Identification of a patient tissue-based phosphorylation site as a biomarker for epidermal growth factor receptor (EGFR) activity in non-small cell lung cancer

#288

Alexander Flohr[1], Nithya Raman[1], Daniel Pohlman[1], Jonathan Woodsmith[1], Judith Schreiber[2], Sarah Y. Weißenberg[2], Anke Nillies[2], Mirja Kastein[2], Malik Khenkhar[2], Kerstin A. David[2], Hartmut Juhl[1]
[1] Indivumed GmbH, Falkenried 88, 20251 Hamburg, Germany; [2] Indivumed Services GmbH, Falkenried 88, 20251 Hamburg, Germany

Indivumed Therapeutics

INTRODUCTION

Non-small cell lung cancer (NSCLC) is a leading cause of cancer-related deaths globally. EGFR tyrosine kinase inhibitors (TKIs) have shown promise in treating NSCLC, yet current biomarkers rely on genetic data alone. Identifying reliable biomarkers that more comprehensively monitor EGFR activity and could predict therapeutic responses may facilitate precision medicine approaches. Here, we report an uncharacterized threonine phosphorylation site within core regulator of the EGFR pathway; SHP2, that is strongly predictive of EGFR activity as defined by genetic mutation status that shows potential as a clinically relevant biomarker.

COHORT OVERVIEW

We performed a multi-omics analysis of NSCLC patients facilitating novel biomarker identification. 977 fresh-frozen, surgically resected tumor and adjacent normal samples were used to generate whole genome sequencing (WGS) and mass spectrometry (whole proteome (WP) and phospho-proteome (PP) profiling) data. For this project the adenocarcinoma results were taken forward for analysis.

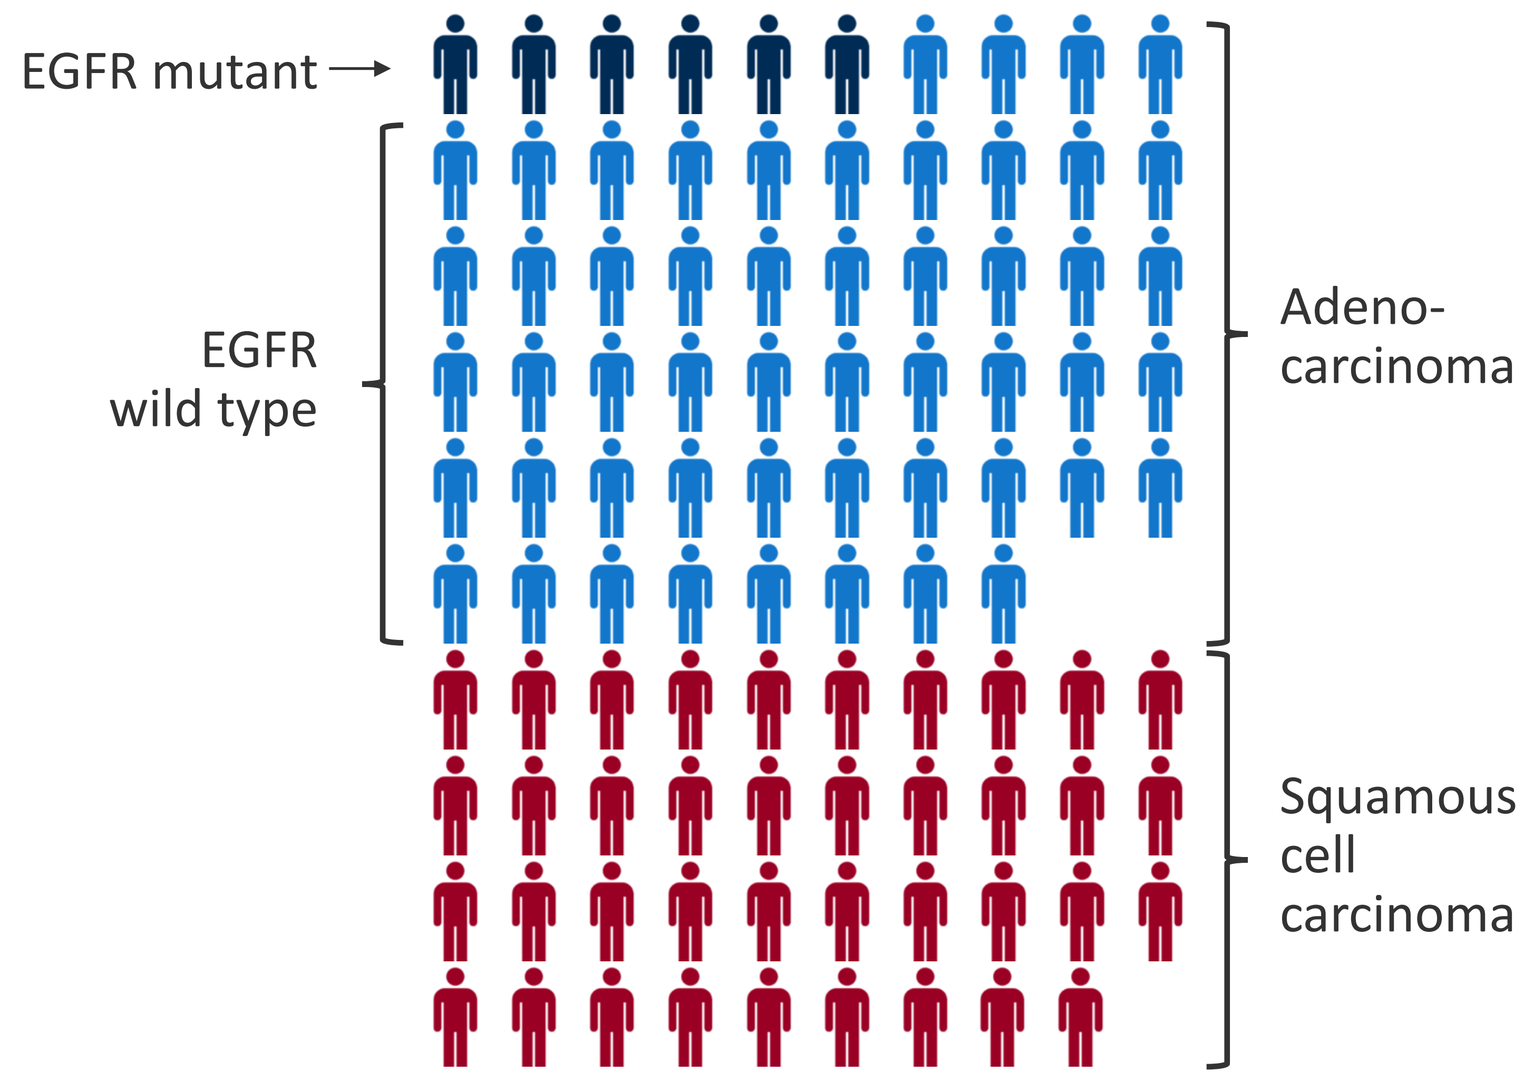
EGFR mutant
EGFR wild type
Adeno-carcinoma
Squamous cell carcinoma

Figure 1: EGFR mutation status classification on the NSCLC cohort based on known constitutive receptor activation mutations using WGS data. Here we observe ~10% of adenocarcinoma cases carrying an activated EGFR.

METHODS

Collection of samples: Tissues were collected using standardized protocols, minimizing cold ischemia time prior to freezing in liquid nitrogen. To ensure sample quality, all tissues were H&E stained and subjected to a pathological QC. Samples need to be invasive, have a tumor content of >= 30 % and Necrosis <= 30 %.

NGS Sequencing Data Collection & Data Processing: After successful QC, approx. 10 mg tissue material is taken for nucleic acid extraction. For DNA, tissues are homogenized using the BeadBug system and DNA extracted using the Qiagen AllPrep Universal Kit. Libraries for (WGS) are prepared using the KAPA Hyper Prep Kit (PCR-free). Sequencing is performed on a NovaSeq6000. NGS data was aligned against Grch38. Small somatic variation was called using a consensus approach from Mutect2 [1], Strelka [2], Varscan [3] and Somatic Sniper [4] callers.

Mass Spectrometry Data Collection: For WP profiling, 5-10 mg of tissue was lysed in Precellys® Evolution homogenizer with ceramic beads & lysis buffer containing PhosSTOP™. Digest peptides were then acidified and desalted. PP samples were generated by phosphopeptide enrichment (MagReSyn Ti-IMAC beads, ReSyn Biosciences) using 500 µg of the WP sample. For DIA LC-MS/MS measurements, 5 µg of peptide samples were loaded into an ACQUITY M-Class micro-flow LC system (Waters) and peptides eluted into a Q Exactive HF-X mass spectrometer (Thermo). Tissue-specific spectral libraries were generated combining DDA and DIA measurements and raw data processed using software Spectronaut v13.

Immunohistochemical analysis: IHC of human Formalin-Fixed Paraffin-Embedded (FFPE) NSCLC tissue samples was performed on the Discovery Ultra staining platform (Ventana/Roche) using different antibodies raised against pT59 in mice (AbMart). Whole slide scans were generated with the Axio Scan Z1 automated slide scanner (Zeiss).

Phospho-proteogenomics

Proteome wide comparison between EGFR wild type and activating mutant freshly frozen adenocarcinoma samples.

Across 3,667 proteins and 26,598 phosphosites we see a clear outlier phosphosite that is upregulated upon EGFR mutation.

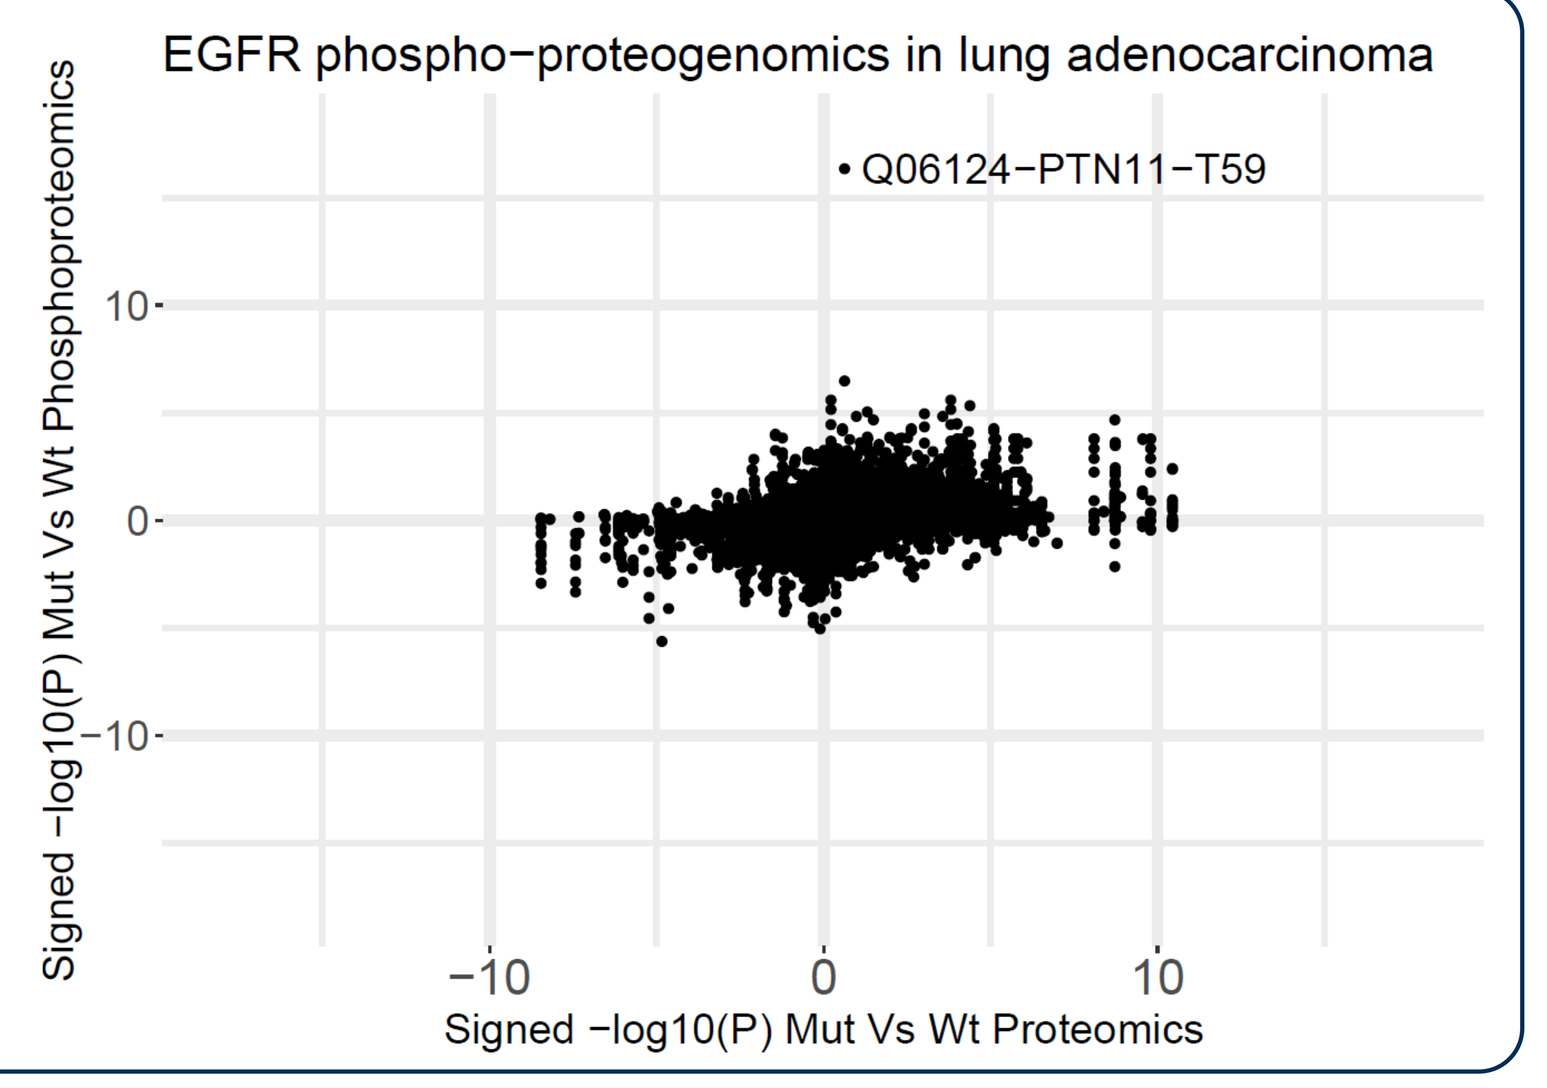
EGFR phospho−proteogenomics in lung adenocarcinoma
Q06124−PTN11−T59
Signed −log10(P) Mut Vs Wt Phosphoproteomics
10
0
−10
−10
0
10
Signed −log10(P) Mut Vs Wt Proteomics

Figure 2: EGFR based phospho-proteogenomics. Only proteins also annotated with phosphorylation sites plotted. Statistics calculated using wilcoxen test, signed using protein/phosphosite up/down regulation.

Patient-based pT59 PTPN11(SHP2) Signal

Importantly, phosphorylation on T59 in PTPN11(SHP2) is not altered in associated normal tissue (Fig 3A).

Individual phosphosite level data is critical, as classifiers built combining the intensities of either all phospho-threonines or all phosphosites on PTPN11 perform worse than the pT59 alone (Fig 3B). Concordantly, a classifier without pT59 performs most poorly.

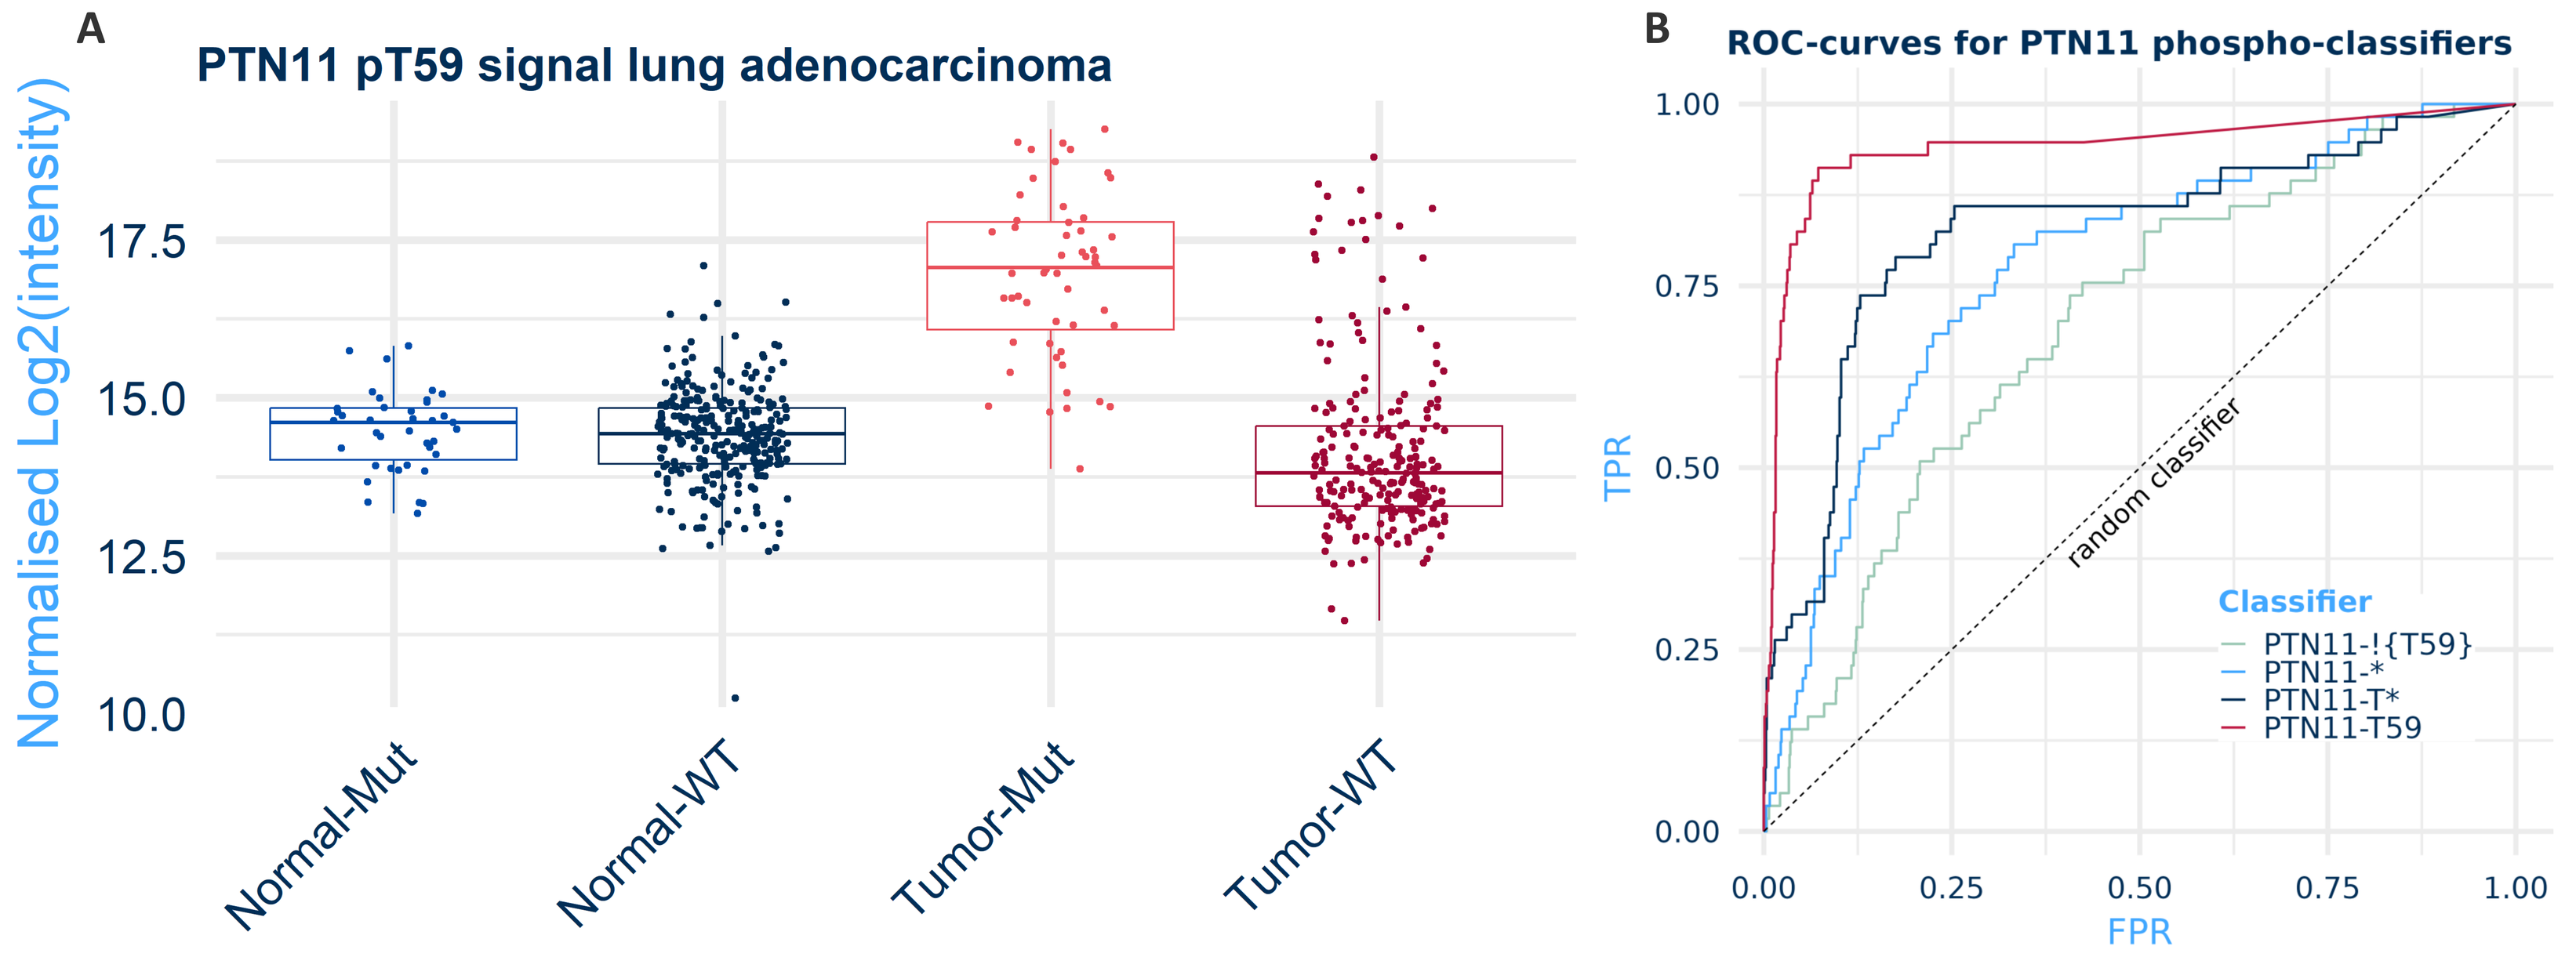
A
PTN11 pT59 signal lung adenocarcinoma
Normalised Log2(intensity)
17.5
15.0
12.5
10.0
Normal-Mut
Normal-WT
Tumor-Mut
Tumor-WT
B
ROC-curves for PTN11 phospho-classifiers
TPR
1.00
0.75
0.50
0.25
0.00
0.00
0.25
0.50
0.75
1.00
FPR
random classifier
Classifier
PTN11-!{T59}
PTN11-*
PTN11-T*
PTN11-T59

Figure 3 A: Distinction between tumor samples carrying an activating mutation in EGFR in comparison to wild type samples, with a comparative analysis in the normal tissue counterparts. **B:** PTPN11-pT59 as a classifier of EGFR activity as defined by genetic mutations reports an AUC of 0.94. Comparative classifiers using all measured phospho-threonine (T*) sites , all phosphorylation sites on PTPN11 (*) or all phosphorylation sites on PTPN11 but pT59 (!{T59}).

Structural Insights into T59

Using a PTPN11(SHP2) structure [6], we observe T59 (highlighted in green, Fig 3) in the D'E loop of the N terminal SH2 domain, known to interacts with the catalytic phosphatase domain WPD-loop.

Phosphorylation of T59 would influence intramolecular bonds within the loop (inset view, Fig 4), likely leading to conformational changes in the SH2 domain that could impact the phosphatase activity.

Interestingly, T59 is also in spatial proximity to the cancer associated T507K mutations [6], as well as a known EGFR responsive phosphosite, Y62 [7].

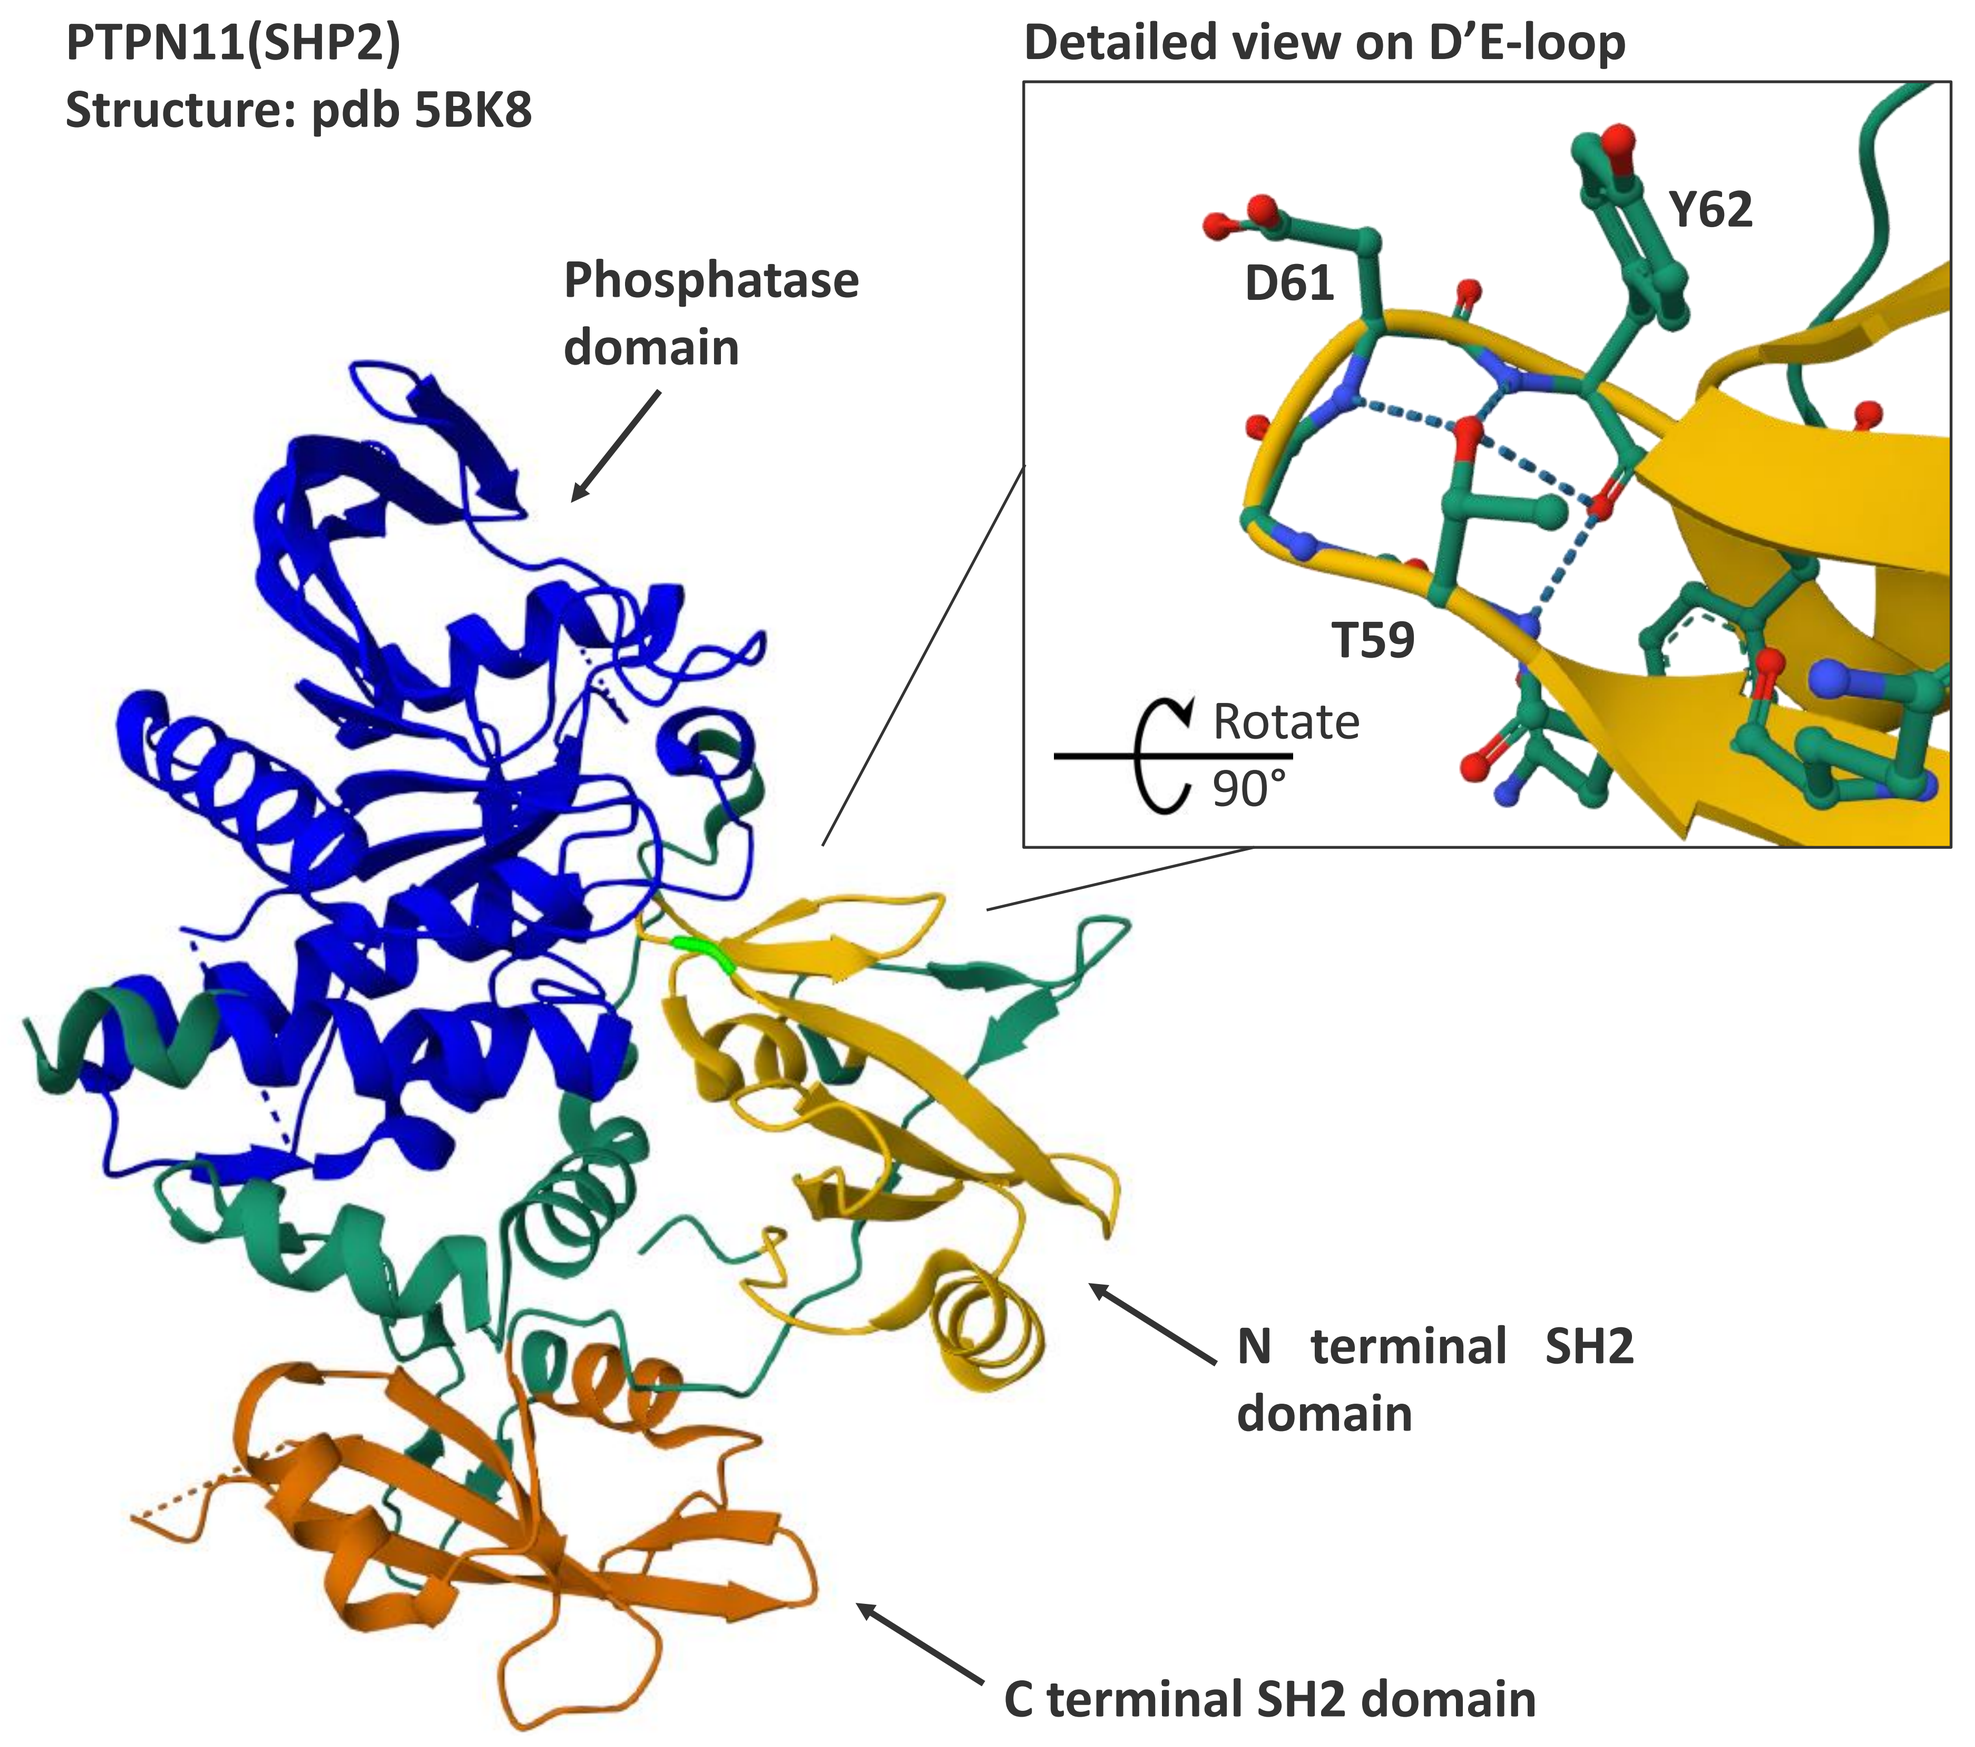
PTPN11(SHP2)
Structure: pdb 5BK8
Detailed view on D'E-loop
Phosphatase domain
Y62
D61
T59
Rotate 90°
N terminal SH2 domain
C terminal SH2 domain

Figure 4: PTPN11 structure highlighting the position of T59 (neon green) at the interface between the SH2 and phosphatase domain. Inset: Detailed view on existing intramolecular bonds mediated by T59 in the D'E-loop within the SH2 domain.

Phosphosite Stability Analysis

An insight into the stability of the phospho-site can be ascertained through the tumor sample cold-ischemia time distribution.

We see no difference in phosphosite intensity across 20 minutes, suggesting a robust phosphosite signal that may be suitable for clinical formalin fixed samples.

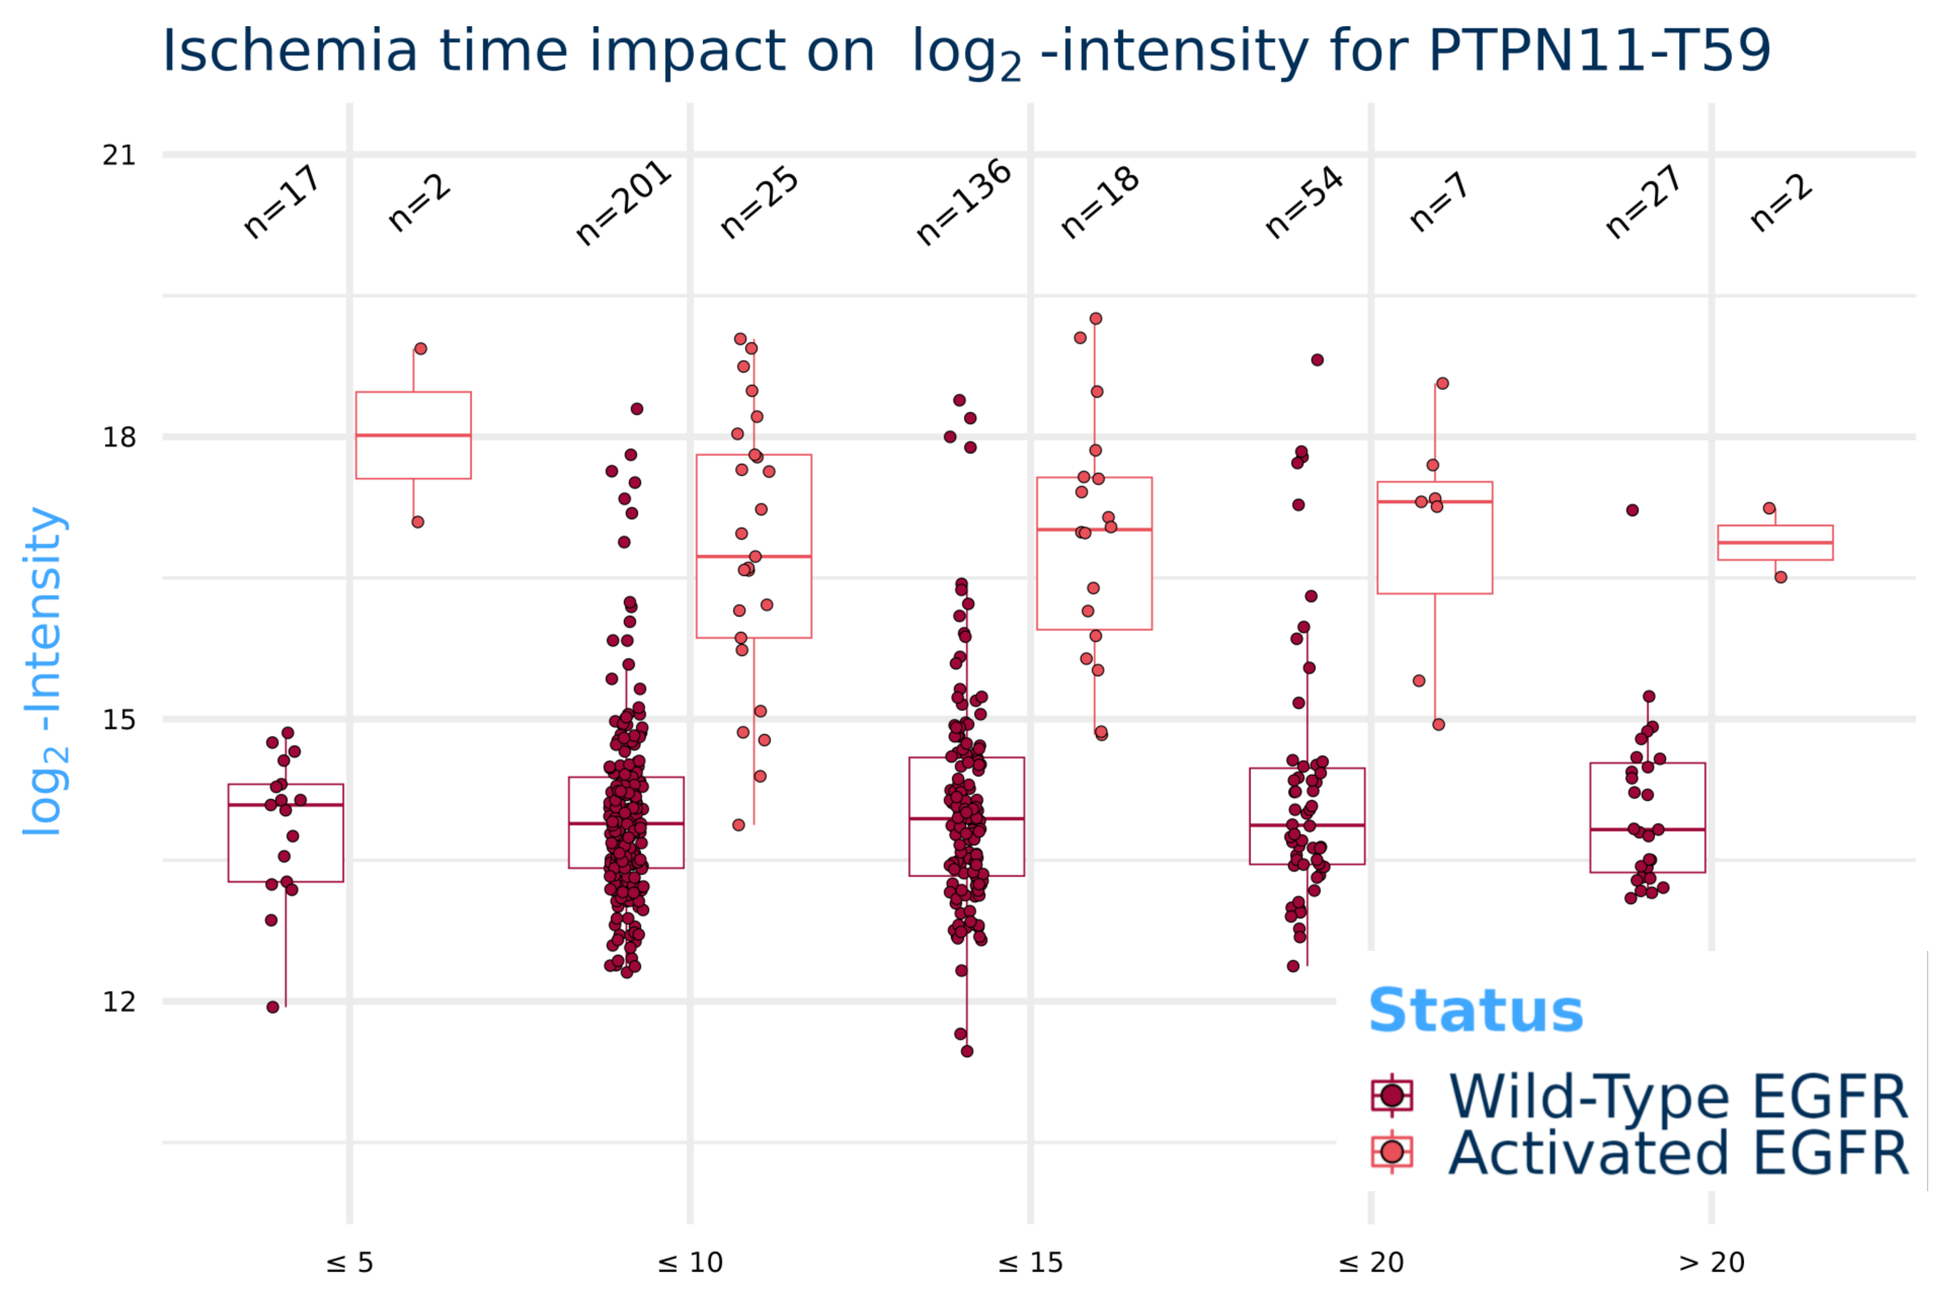
Ischemia time impact on log2 -intensity for PTPN11-T59
log2 -Intensity
21
18
15
12
n=17
n=2
n=201
n=25
n=136
n=18
n=54
n=7
n=27
n=2
≤ 5
≤ 10
≤ 15
≤ 20
> 20
Status
Wild-Type EGFR
Activated EGFR

Figure 5: PTPN11 pT59 characteristics across cold-ischemia time within the lung adenocarcinoma sample set.

FFPE-tissue Antibody Validation of pT59 Signal

Monoclonal antibodies were generated against a PTPN11(SHP2) T59 phosphorylated peptide. These antibodies showed a good discriminatory signal in comparison to the unphosphorylated peptide in ELISA assays (AbMart, data not shown).

The antibodies were then tested on standard FFPE tissues in immuno-histochemical (IHC) assays.

Wild type EGFR lung adenocarcinoma samples that showed no pT59 signal in mass spec data also showed little signal in the IHC experiments (Fig 6A).

Conversely, the pT59 mass spec signals were pheno-copied in EGFR mutant lung adenocarcinoma samples (Fig 6B), with a strong staining in the IHC assay.

More extensive testing needs to be carried out to comprehensively assay how well the antibody mirrors the mass spec fresh frozen tissue results.

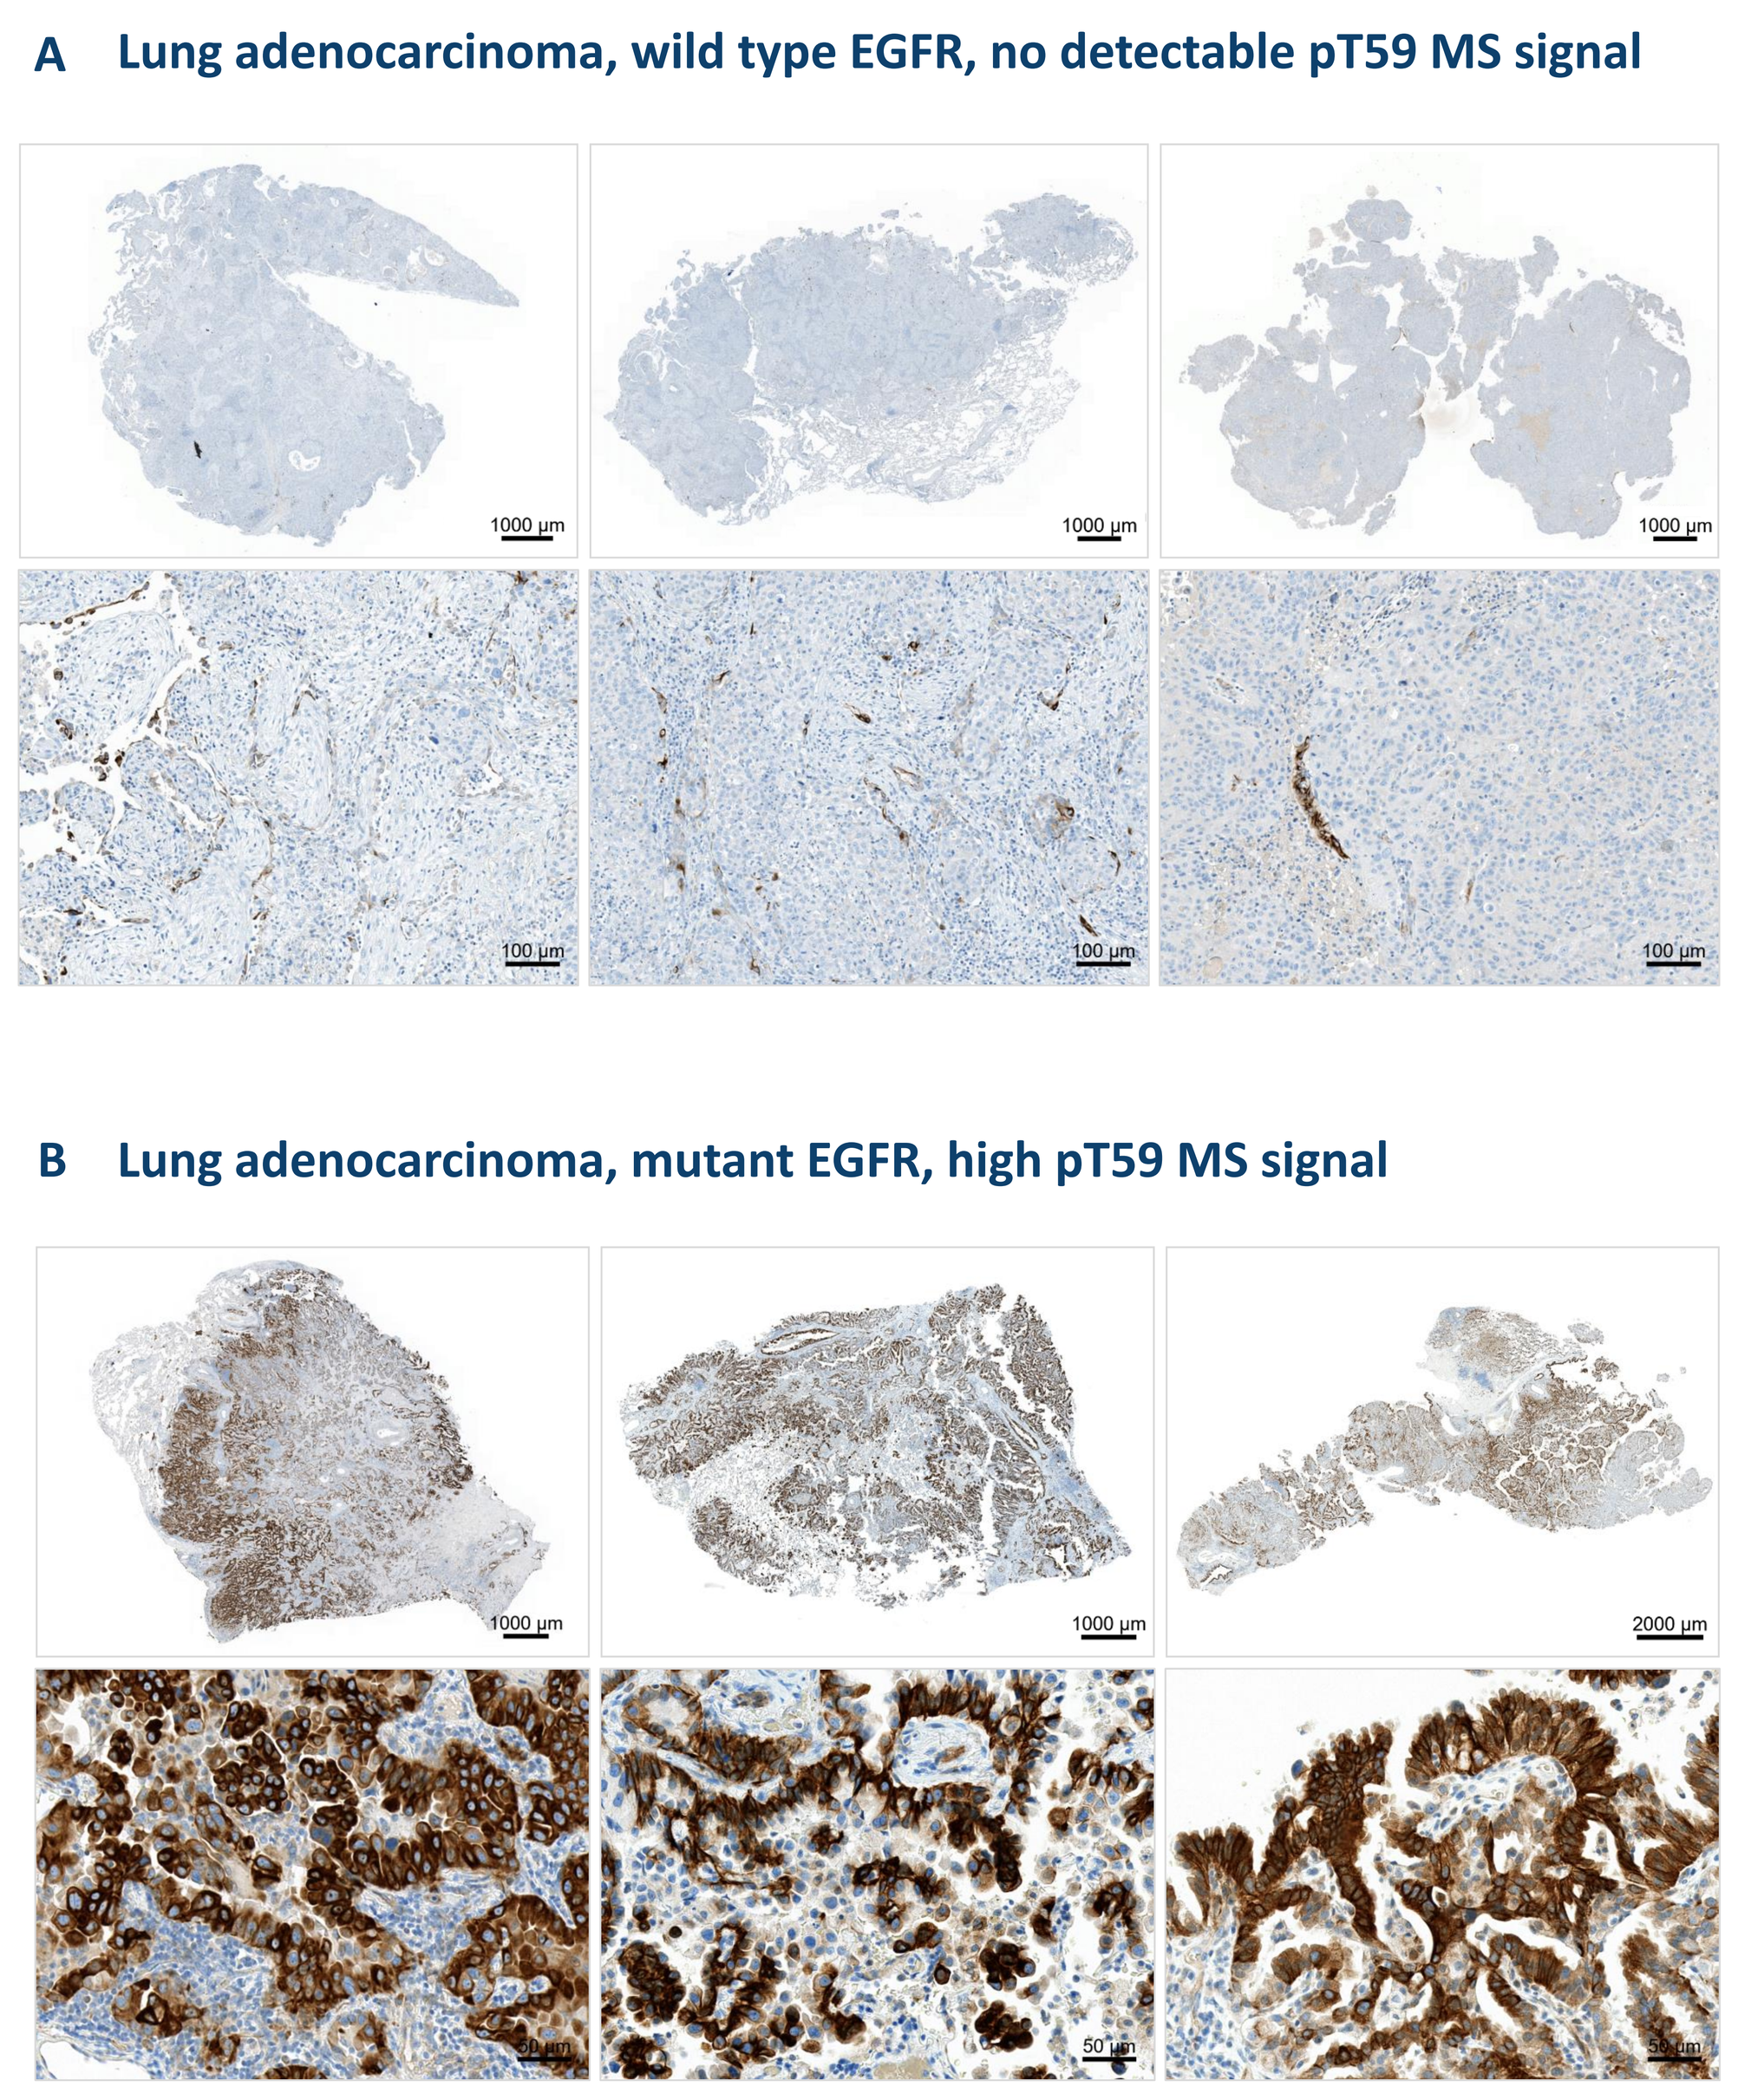
A Lung adenocarcinoma, wild type EGFR, no detectable pT59 MS signal
1000 µm
1000 µm
1000 µm
100 µm
100 µm
100 µm
B Lung adenocarcinoma, mutant EGFR, high pT59 MS signal
1000 µm
1000 µm
2000 µm
50 µm
50 µm
50 µm

Figure 5: IHC assay testing a monoclonal antibody against pT59 in PTPN11(SHP2) in lung adenocarcinoma FFPE tissue from **A** wild type EGFR and **B** mutant EGFR.

CONCLUSION

Our findings suggest that this uncharacterized SHP2 phosphosite may serve as a potent biomarker for EGFR activity in lung adenocarcinoma. Further validation of the monoclonal antibodies as tools for this clinically relevant biomarker is required in larger patient cohorts to further investigate the pT59 signal in comparison to EGFR activation and to elucidate the role of the phosphosite in therapy response. This novel marker has the potential to guide therapeutic decision-making and contribute to personalized medicine strategies for NSCLC patients.

REFERENCES

1. van der Auwera, G. et al.: *Genomics in the Cloud: Using Docker, GATK, and WDL in Terra.* O'Reilly Media, 2020, Incorporated. ISBN: 9781491975190
2. Kim, S., et al.*: Strelka2: fast and accurate calling of germline and somatic variants. Nat Methods*, 2018. 15(8): p. 591-594.
3. Koboldt, D. C., et al.: *VarScan: variant detection in massively parallel sequencing of individual and pooled samples.* Bioinformatics 2009. 25(17): p. 2283-5.
4. Larson, D. E. et al.: *SomaticSniper: identification of somatic point mutations in whole genome sequencing data*. Bioinformatics, 2012, 28(3):311-317
5. Bruderer, R. et al.: *Analysis of 1508 plasma samples by capillary flow data-independent acquisition profiles proteomics of weight loss and maintenance*. MCP 18 (6), 1242-1254 (2019)
6. Zhang, RY et al.: Mechanistic insights explain the transforming potential of the T507K substitution in the protein-tyrosine phosphatase SHP2 J. Biol. Chem. 295(18) 6187–6201. (2020)
7. Gillette MA et al.: Proteogenomic Characterization Reveals Therapeutic Vulnerabilities in Lung Adenocarcinoma. Volume 182, Issue 1, 9 July 2020, p200-225.e35