Abstract Presentation Number # 6775

Combination of Spatial Transcriptomics and Multiplex IHC as Valuable Multi-Omics Tool for Biomarker Discovery

Nicole Kerstedt, Monika Schoeppler, Bita Motamedi-Baniassad, Antonia Hirt, Julia Ploetzky, Mirja Kastein, Laura Thoms, Malik Khenkhar, Hartmut Juhl, and Kerstin A. David

INTRODUCTION

Colorectal cancer (CRC) is one of the most common cancers worldwide and shows heterogenous molecular subtypes making personalized medicine as well as identifying new targets highly important to provide individual patients with the most appropriate treatment.

To gain more insight into the visualization of potential molecules or pathways for targeting CRC we used a multi-omics approach for the simultaneous analysis of gene and protein expression within a sample. In this study we combined fluorescent multiplex immunohistochemistry (mIHC) for the detection of proteins (3-plex panel & DAPI) with the Visium Spatial Transcriptomics technology (10x Genomics) that enables the detection of the whole transcriptome in the histological context.

METHODS

Samples: Biospecimen were collected immediately after resection according to Indivumed's standard operating procedures. An informed consent was obtained from patients. For the study, a formalin-fixed paraffin-embedded (FFPE) tumor tissue sample of one patient diagnosed with CRC stage 1 was selected.

Tissue Sectioning: The FFPE tumor tissue block was sequentially sectioned using a microtome: One 3 µm section for mIHC staining, one 3 µm section for hematoxylin and eosin (H&E) staining followed by one section of 5 µm for the Visium Spatial Transcriptomics workflow.

mIHC: Tyramide signal amplification (TSA) based sequential fluorescent 4-color mIHC (p53/ p21/ Ki-67 + DAPI) on one FFPE tissue section was performed on the Leica BOND RX staining platform using the following antibody fluorophore combinations and order: Anti-p53 clone DO-7 (Roche/ Ventana) and Opal 520 (Akoya Biosciences), anti-p21 clone DCS-60.2 (Roche/ Ventana) and Opal 650 (Akoya Biosciences), anti-Ki-67 clone 30-9 (Roche/ Ventana) and Opal 570 (Akoya Biosciences), as well as DAPI as counterstain. Scans were generated with the Axio Scan.Z1/ ZEN 3.5 automated slide scanner/ software (Zeiss).

10x Genomics Visium Spatial Gene Expression Manual Workflow for FFPE (v1): One FFPE section was mounted on a Visium Spatial Gene Expression Slide containing spatially barcoded capture primers. The FFPE section was first H&E stained and scanned to visualize the histological tissue structure, followed by human whole transcriptome probe hybridization, targeting 17,943 human protein coding genes by 18,630 probe pairs. According to hybridization probes were ligated, followed by tissue permeabilization and probe release. Ligated probes were than captured via spatially barcoded primers and extended followed by Visium Spatial Gene Expression FFPE Library Preparation. The final library was sequenced at a concentration of 300 pM on a NovaSeq™6000 using a S1 v1.5 flowcell (Illumina) to reach a minimum sequencing depth of 25,000 read pairs per tissue covered spot by our sub-provider (IMGM Laboratories). Sequencing data containing the specific spatially sequencing barcodes were analyzed using the Space Ranger analysis pipeline (version 1.3.0) and the Loupe Browser (version 6.4.1) to assign the gene expression data to the corresponding histological positions in the tissue section (Figure 1).

Tissue Preparation	Sample Processing, Imaging, Library Preparation		Sequencing	Data Processing
• Tissue preparation • FFPE Tissue Sectioning • Placement of tissue section on Gene Expression Slide	• Deparaffinization • H&E staining • Brightfield Imaging with Zeiss Axio Scan.Z1 • Decrosslinking	• Human whole transcriptome probe hybridization & Ligation • Permeabilization & Capturing • Library construction & Library QC	• NovaSeq™ 6000 with S1 v1.5 flowcell (Illumina) • Asymmetric paired-end sequencing (28;10;10;90)	• Space Ranger pipelines: processing of RNA-seq data and H&E image • Loupe Browser: providing interactive visualization

Figure 1: Visium Spatial Gene Expression Manual Workflow for FFPE (v1) (10x Genomics).

Experimental Setup

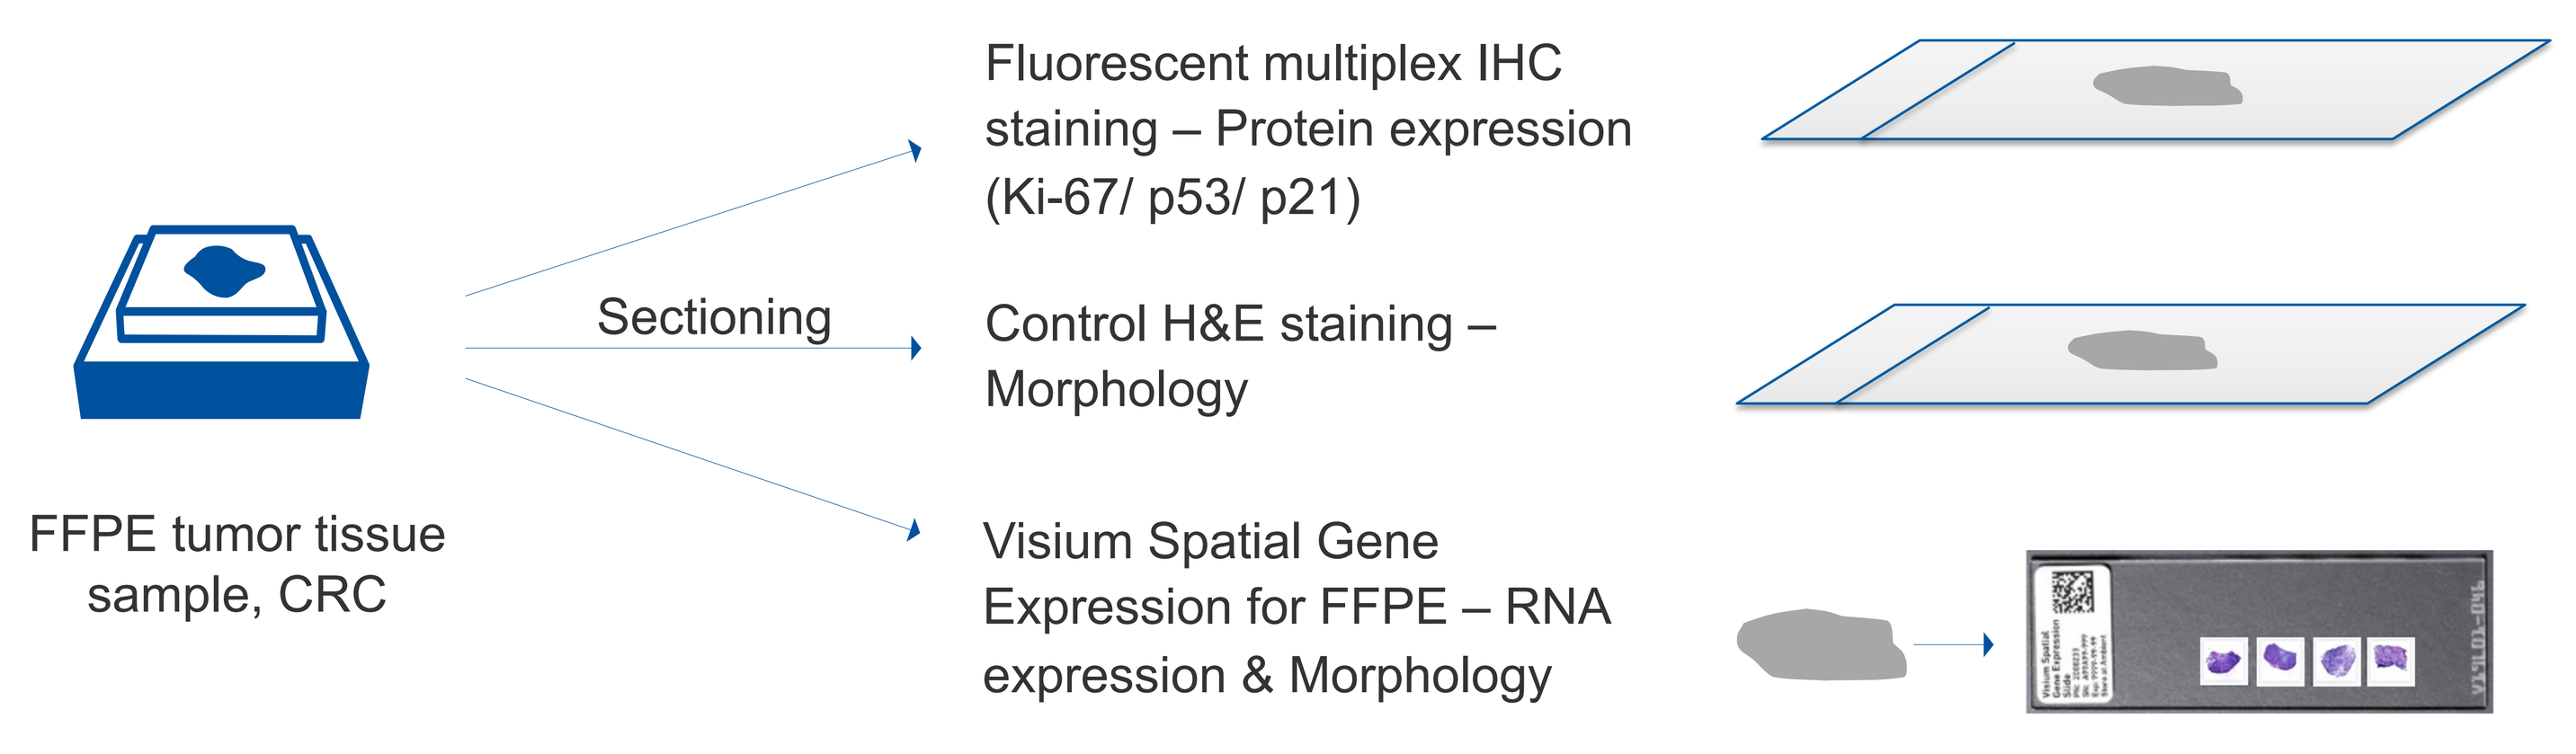
FFPE tumor tissue sample, CRC
Sectioning
Fluorescent multiplex IHC staining – Protein expression (Ki-67/ p53/ p21)
Control H&E staining – Morphology
Visium Spatial Gene Expression for FFPE – RNA expression & Morphology

Figure 2: Experimental setup. One FFPE tumor tissue block of a patient diagnosed with CRC was sequentially sectioned for a multi-omics approach. The first 3 µm section was used for fluorescent mIHC staining visualizing the protein expression of Ki-67 (proliferation marker), p53 and p21 (both tumor suppressor genes). The second 3 µm section was stained for hematoxylin and eosin (H&E) to evaluate the tissue morphology. On the third 5 µm section the 10x Genomics Visium Spatial Gene Expression manual workflow for FFPE was applied, and gene expression patterns were analyzed by using the software Space Ranger and Loupe Browser.

RESULTS – Multi-Omics Data

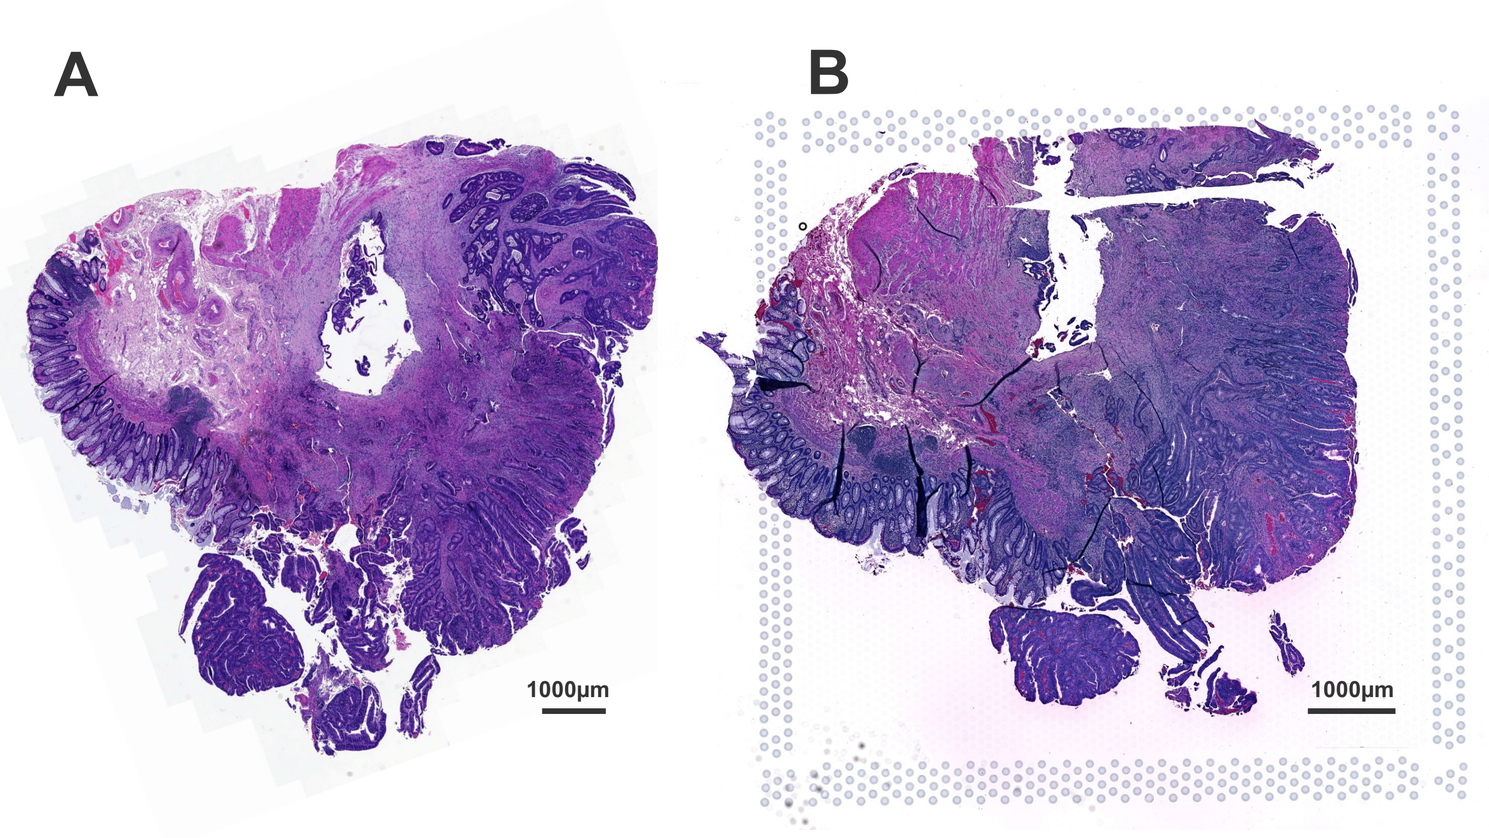
A
B
1000µm
1000µm

Figure 3: mIHC, H&E stain and Visium data. **A:** Control H&E staining and **B:** Visium H&E staining showing the morphology of the CRC tumor tissue sample. **C-F:** Fluorescent mIHC protein detection with anti-Ki-67, anti-p53 and anti-p21 staining (orange, green, red) as well as an overlay including nuclei staining (DAPI, blue). **G-J:** Visium Spatial Gene Expression images of MKI67, TP53 and CDKN1A as well as an overlay of the expression profiles of the three genes within the tumor tissue sample. Gene expression is shown in Log2 expression.

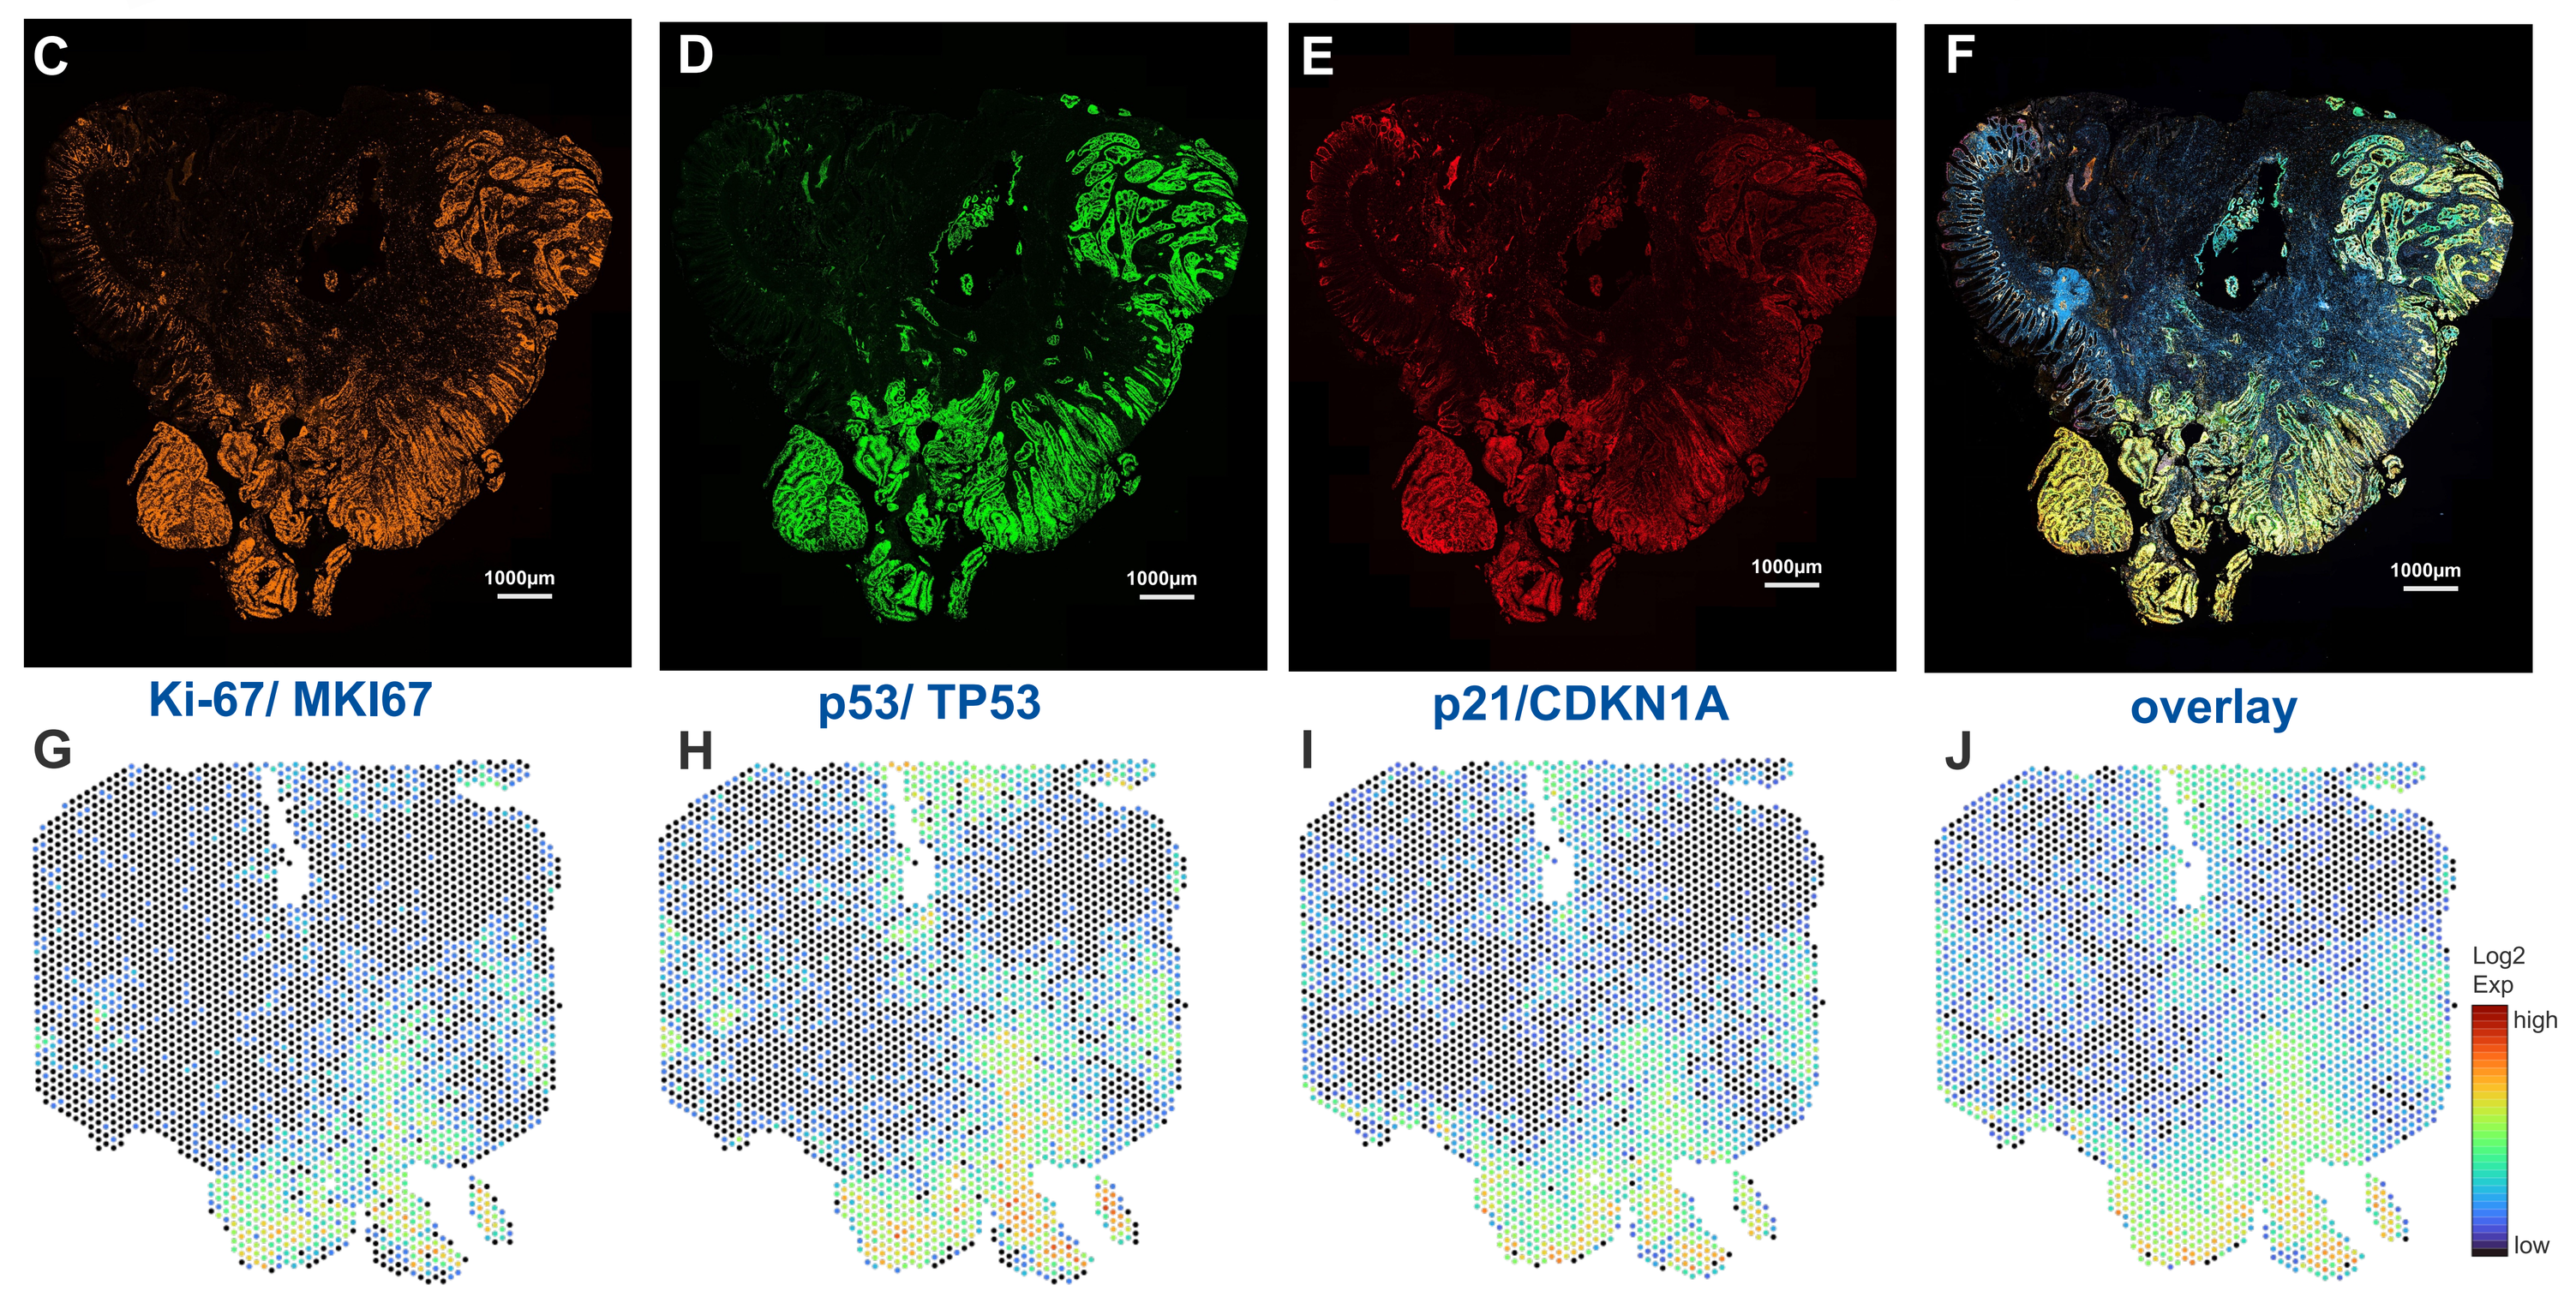
C
D
E
F
1000µm
Ki-67/ MKI67
p53/ TP53
p21/CDKN1A
overlay
G
H
I
J
Log2 Exp
high
low

Good correlations of protein and gene expression of Ki-67, p53 and p21 were observed within the analyzed CRC tumor tissue sample. Furthermore, clear differences of the expression patterns were detectable between tumor cell areas and regions with corresponding normal mucosa cells.

RESULTS – Discovery of Potential New Markers

By comparing Spatial Transcriptomics gene expression patterns of tumor tissue and corresponding normal adjacent tumor tissue, we identified genes that are associated with the p53-pathway, e.g. proline rich acidic protein 1 (PRAP1)[1]. Moreover, we identified genes that were only poorly described in cancer, e.g. Serin Protease 33 (PRSS33) or genes that have been linked to other cancer types, with only little known about their role in CRC, e.g. Suppressor APC domain containing 2 (SAPCD2)[2]. Corresponding protein expression can be verified in the next steps by using different protein assays such as chromogenic IHC.

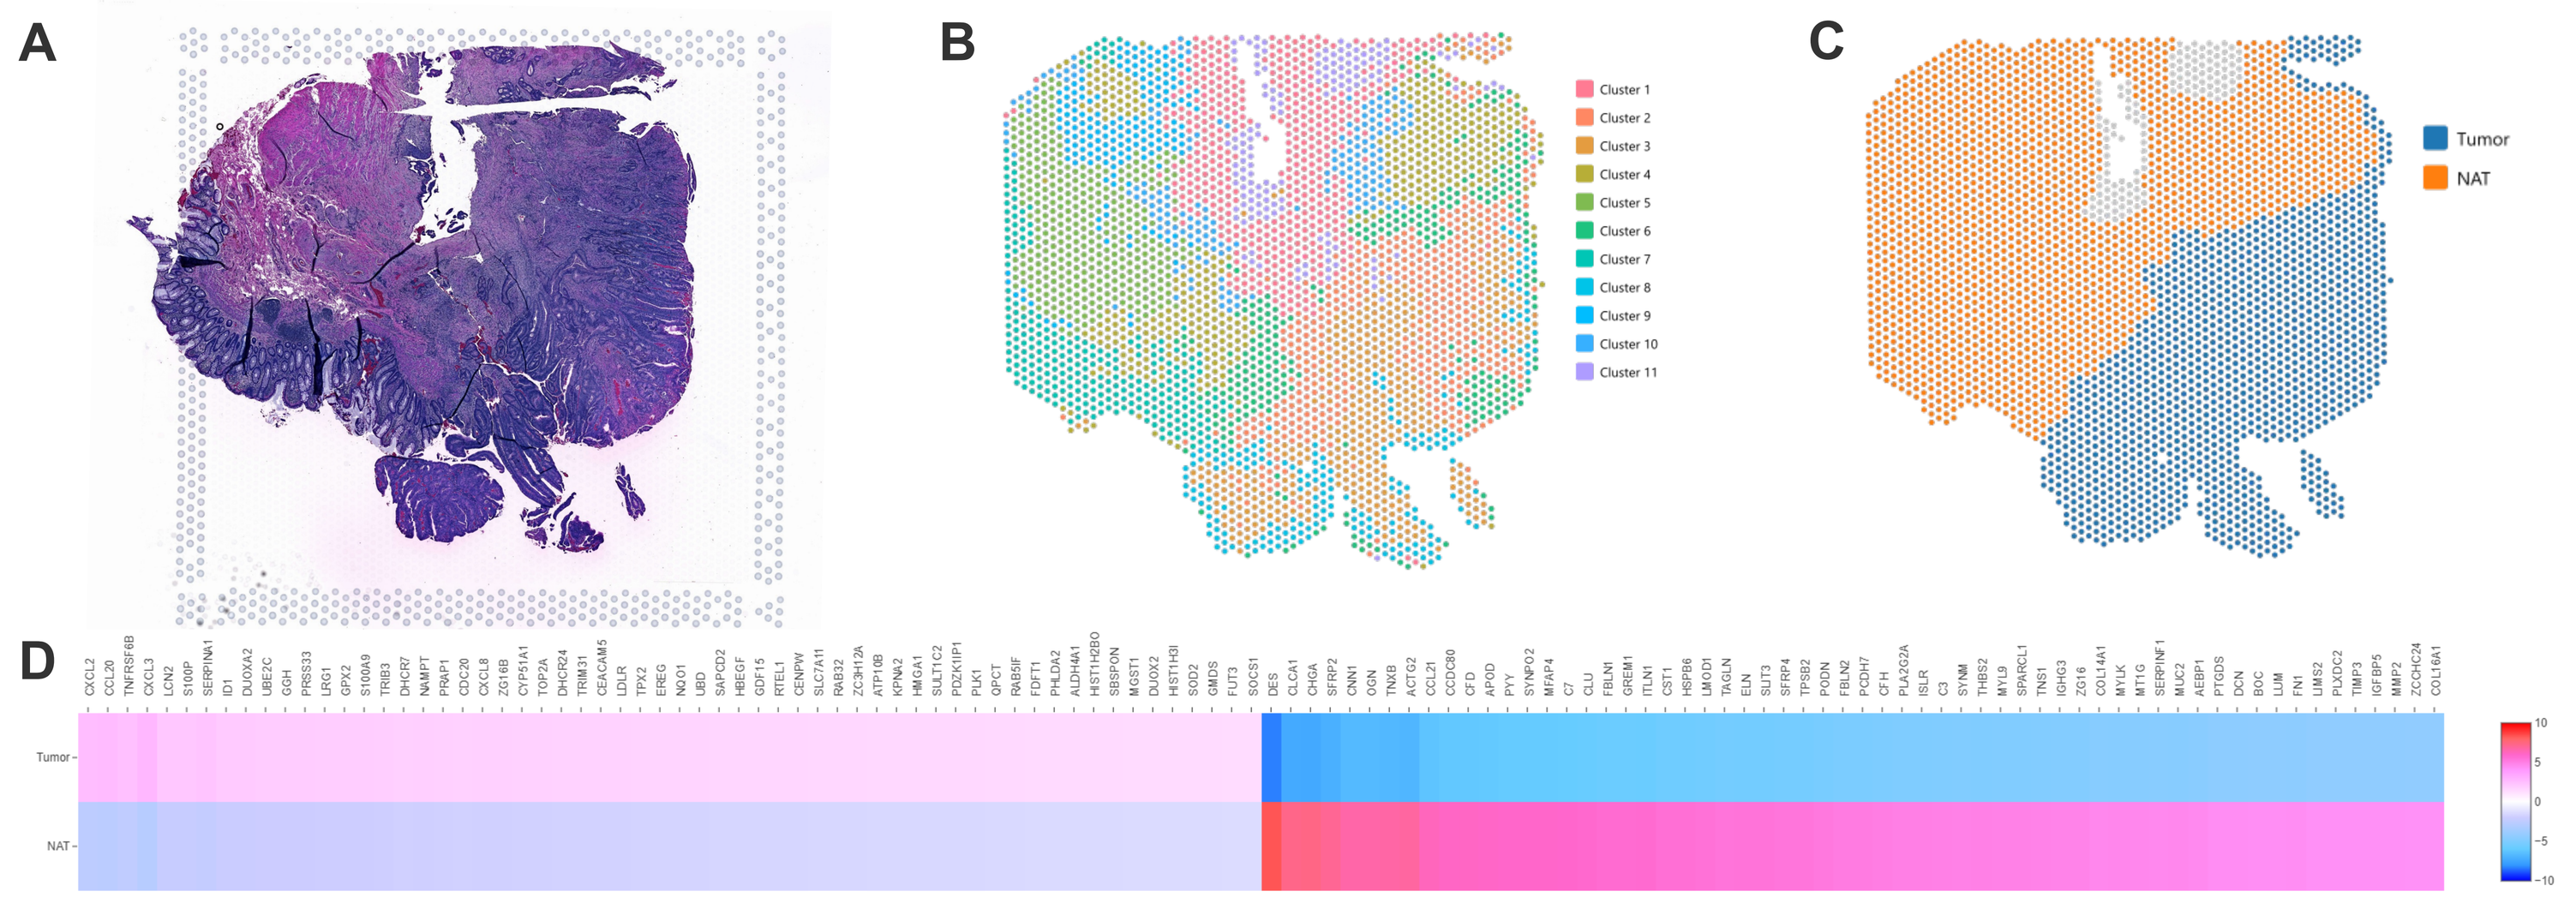
A
B
C
D
Cluster 1
Cluster 2
Cluster 3
Cluster 4
Cluster 5
Cluster 6
Cluster 7
Cluster 8
Cluster 9
Cluster 10
Cluster 11
Tumor
NAT

Figure 4: Evaluation strategy of Visium Gene Expression data: **A:** Visium H&E staining showing the morphology. **B:** Graph-based clustering with 11 clusters was performed after reducing dimensionality of data by Principal Component Analysis (PCA). **C:** Reduction of Graph-based clustering into two clusters reflecting morphology – tumor cell area (Tumor) and corresponding normal adjacent tumor (NAT) tissue comprising e.g. mucosa and submucosa. **D:** Heatmap of graph-based Log2 fold changes showing differentially regulated genes between Tumor and NAT clusters.

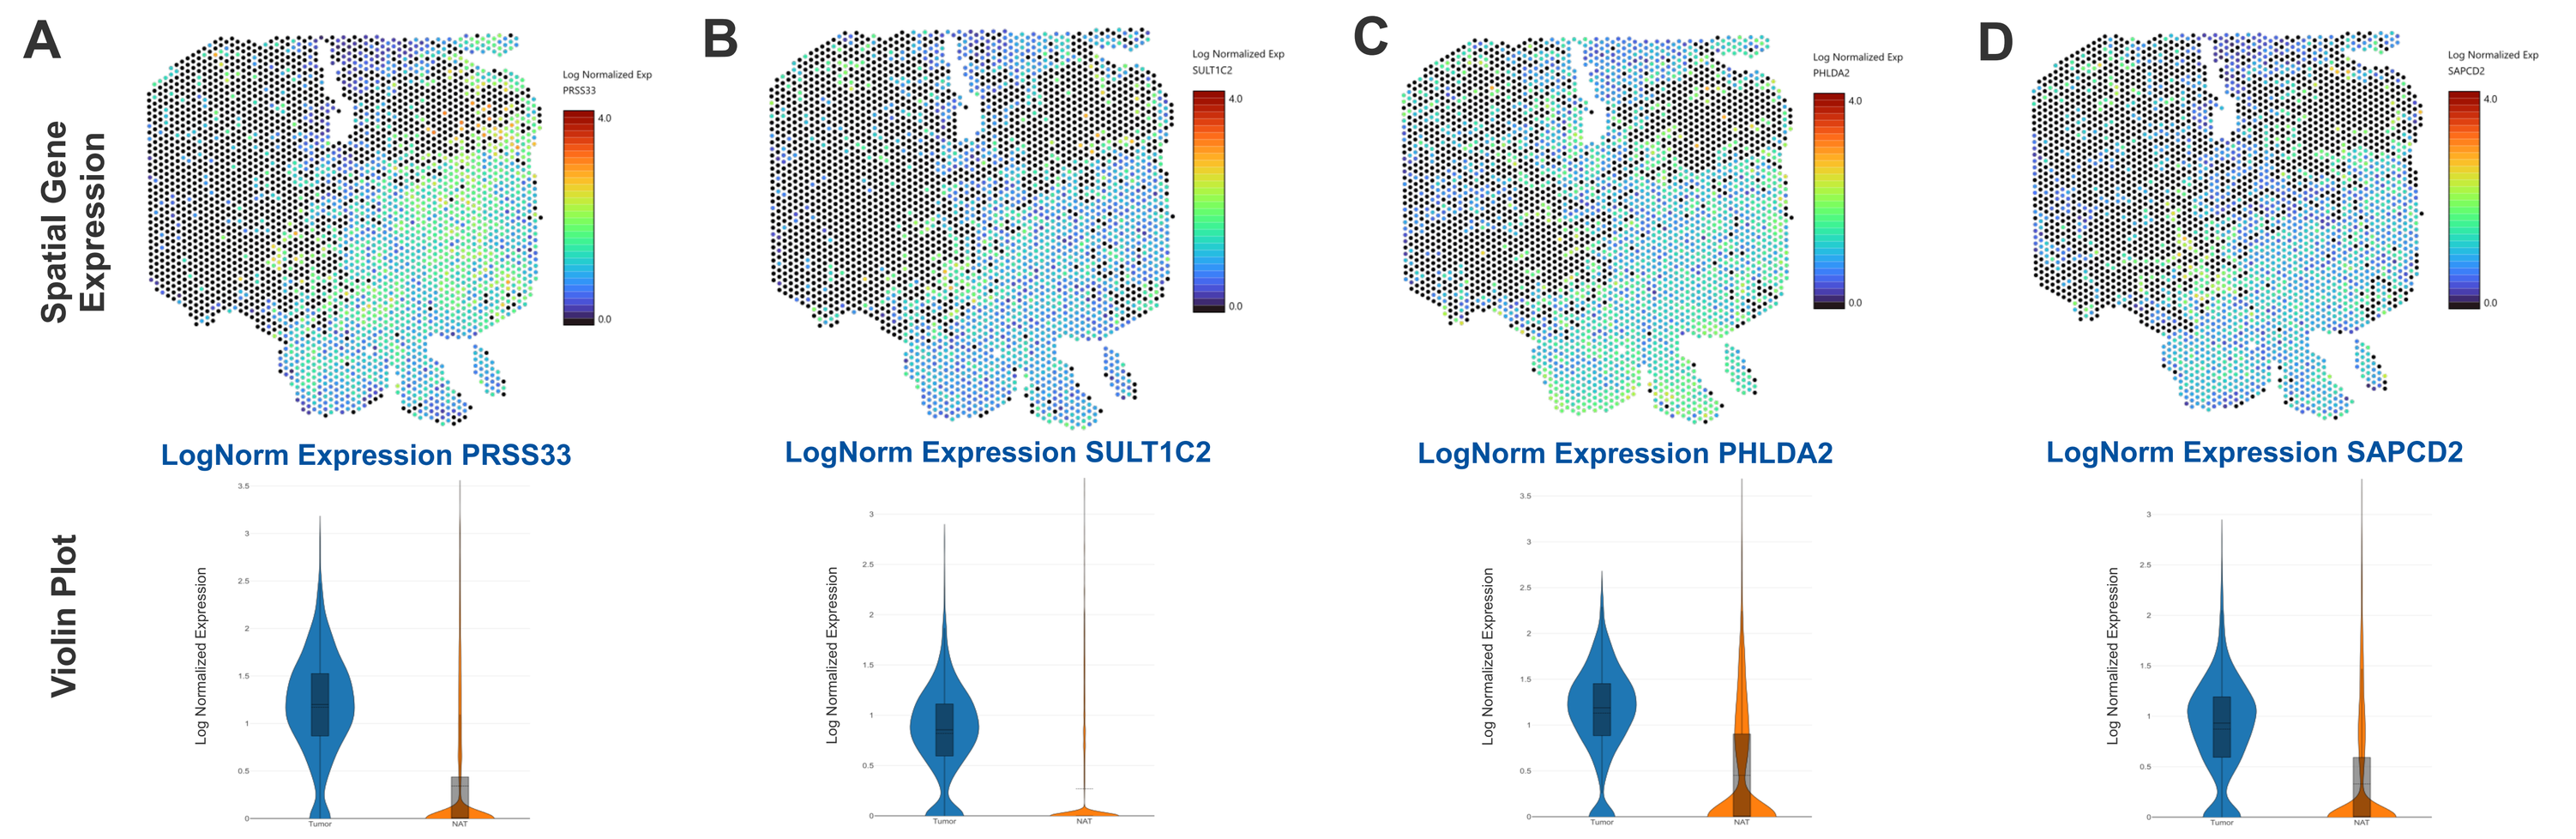
A
B
C
D
Spatial Gene Expression
Violin Plot
LogNorm Expression PRSS33
LogNorm Expression SULT1C2
LogNorm Expression PHLDA2
LogNorm Expression SAPCD2

Figure 5: Spatial Gene Expression of selected significantly up-regulated genes in the analyzed tumor cell area (Tumor) compared to the corresponding normal adjacent tumor (NAT) tissue: Spatial Gene Expression as well as Violin Plots of **A:** Serin Protease 33 (PRSS33). **B:** Sulfotransferase family 1C member 2 (SULT1C2) **C:** Pleckstrin homology like domain family A member 2 (PHLDA2) and **D:** Suppressor APC domain containing 2 (SAPCD2). Spatial Gene Expression and Violine Plots are shown in LogNorm fold changes, reflecting gene expression normalized by UMI counts.

CONCLUSION

The combination of fluorescent mIHC and Spatial Transcriptomics has been shown to be a valuable approach not only to visualize already described relations as proof of principle as demonstrated for p53 and p53-associated genes, but more importantly to enable the discovery of new biomarkers or pathways predominantly linked to CRC tissue that may serve as new targets for cancer treatment.

References:
1. Huang, B H et al. "PRAP1 is a novel executor of p53-dependent mechanisms in cell survival after DNA damage." Cell death & disease vol. 3,12 e442. 13 Dec. 2012
2. Luo, Yage et al. "Overexpression of SAPCD2 correlates with proliferation and invasion of colorectal carcinoma cells." Cancer Cell Int. 2020 Feb 6;20:43